Treatment pathways and efficacy outcomes of patients with metastatic colorectal cancer (mCRC): a real-world prospective, longitudinal, cohort study (PROMETCO)

[1]Andrey Mitroshkin, [2]Jean-Baptiste Bachet, [23]Kalena Marti, [4]Carmine Pinto, [5]György Bodoky, [6]Rocio Garcia-Carbonero, [7]Elias Choucair, [7]Bénédicte Chevallier, [8]Adam Sullivan, [9]Miriam Koopman.

[1]Klinikum Freudenstadt, AkademischesLehrkrankenhaus der UniversitätTübingen, Karl-von-Hahn-Strasse, 100, 72250 Freudenstadt, Germany; [2]Sorbonne Université, Service d'hépato-Gastro-Entérologie, Groupe Hospitalier Pitié Salpêtrière, APHP, Paris, France; [3]Department of Medical Oncology, The Christie NHS Foundation Trust, Wilmslow Road, Manchester, M20 4BX, UK; [4]Medical Oncology, Comprehensive Cancer Centre, AUSL – IRCCS di Reggio Emilia – Viale Risorgimento, 80 42123 Reggio Emilia, Italy; [5]Dél-Pesti Centrumkórház Szent László Telephely Albert Flórián út 5–7 1097 Budapest, Hungary; [6]Oncology Department, Hospital Universitario Doce de Octubre, Imas12, Departamento de Medicina, Universidad Complutense de Madrid (UCM), Avenida De Córdoba s/n, 28041 Madrid, Spain; [7]Servier, 35 rue Verdun, 92284 Suresnes, France; [8]Servier Pharmaceuticals, 200 Pier 4 Blvd, Boston, MA 02210, USA; [9]Department of Medical Oncology, University Medical Centre Utrecht, Utrecht University Heidelberglaan 100 3584 CX Utrecht, the Netherlands.

Presented at ESMO Congress,
October 17-21, 2025
Berlin, Germany

Final publication number: 848P

INTRODUCTION

- Data on real-world treatment of metastatic colorectal cancer (mCRC) in third-line (3L) and beyond is limited to specific agents or to a single country.[1,2]
- With recent advances in mCRC therapies, such as trifluridine/tipiracil (FTD/TPI) ± bevacizumab, or fruquintinib, 3L treatment is no longer considered rescue therapy with prolonged survival benefits being observed in clinical trials.[3, 4] However, there is little information on real-world treatment patterns or the impact of these treatment patterns, or other variables, on survival and other outcomes.
- PROMETCO (NCT03935763) is the first international, prospective, real-world study of treatment in patients with mCRC after two disease progressions since diagnosis.

AIMS

To assess real-world treatment patterns and effectiveness in patients with mCRC

METHODS

- Enrolment in PROMETCO started in March 2019 and all eligible patients at recruiting centres were included.[5]
- Inclusion criteria were: ≥18 years of age, confirmed diagnosis of mCRC, two disease progressions, and willingness to receive subsequent treatment.[5]
- Data for lines of therapy (LoTs) were collected retrospectively before two disease progressions and prospectively during follow-up, including effectiveness data.
- Follow-up was regular, but with no fixed schedule, over an 18-month period.
- The 4 most frequently used treatment sequences were identified based on number of patients of each treatment group per line.
- Treatment patterns and median OS from first-line (1L) (mOS; calculated from 1L to remove bias of patients with pre-1L surgery) is presented here.
- A multivariable analysis was carried out from 1L and from inclusion in PROMETCO to assess variables impacting mOS.

TAKE-HOME MESSAGES

- **Patients received treatment sequences in line with guidelines, with use of chemotherapy (CT) and biologic agents in 1L and 2L, and FTD/TPI in 3L.**
- **Best mOS was observed when patients sequentially received CT + anti-VEGF/anti-EGFR in 1L/2L and 3L FTD/TPI ± bevacizumab, but selection bias cannot be excluded.**
- **Only a few patients received FTD/TPI + bevacizumab due to the timing of this study, however efficacy in these patients was consistent with the results of SUNLIGHT.[3]**
- **PROMETCO results in the real-world setting confirm the use of FTD/TPI monotherapy in 3L, paving the way to the use of FTD/TPI + bevacizumab as the current SOC in the 3L setting as recommended by ESMO guidelines (level [I,A] MCBS 4).**

RESULTS

Treatment pathway

- Overall, 736 patients were included in PROMETCO: 608 patients received ≥3 LoTs, and 400 patients received 3 LoTs and did not proceed to fourth-line (4L) treatment. Treatment sequences according to biomolecular status aligned with recommendations from ESMO.
- The most frequently used treatments at each LoT were 1L chemotherapy (CT) + biologic (505 patients, 68.6%), second-line (2L) chemotherapy (CT) + biologic (418 patients, 58.3%), 3L trifluridine/tipiracil (FTD/TPI) ± bevacizumab (402 patients, 66.1%), and 4L regorafenib (87 patients, 41.8%) (**Figure 1**).

Figure 1: Sankey diagram showing first 4 lines of treatment

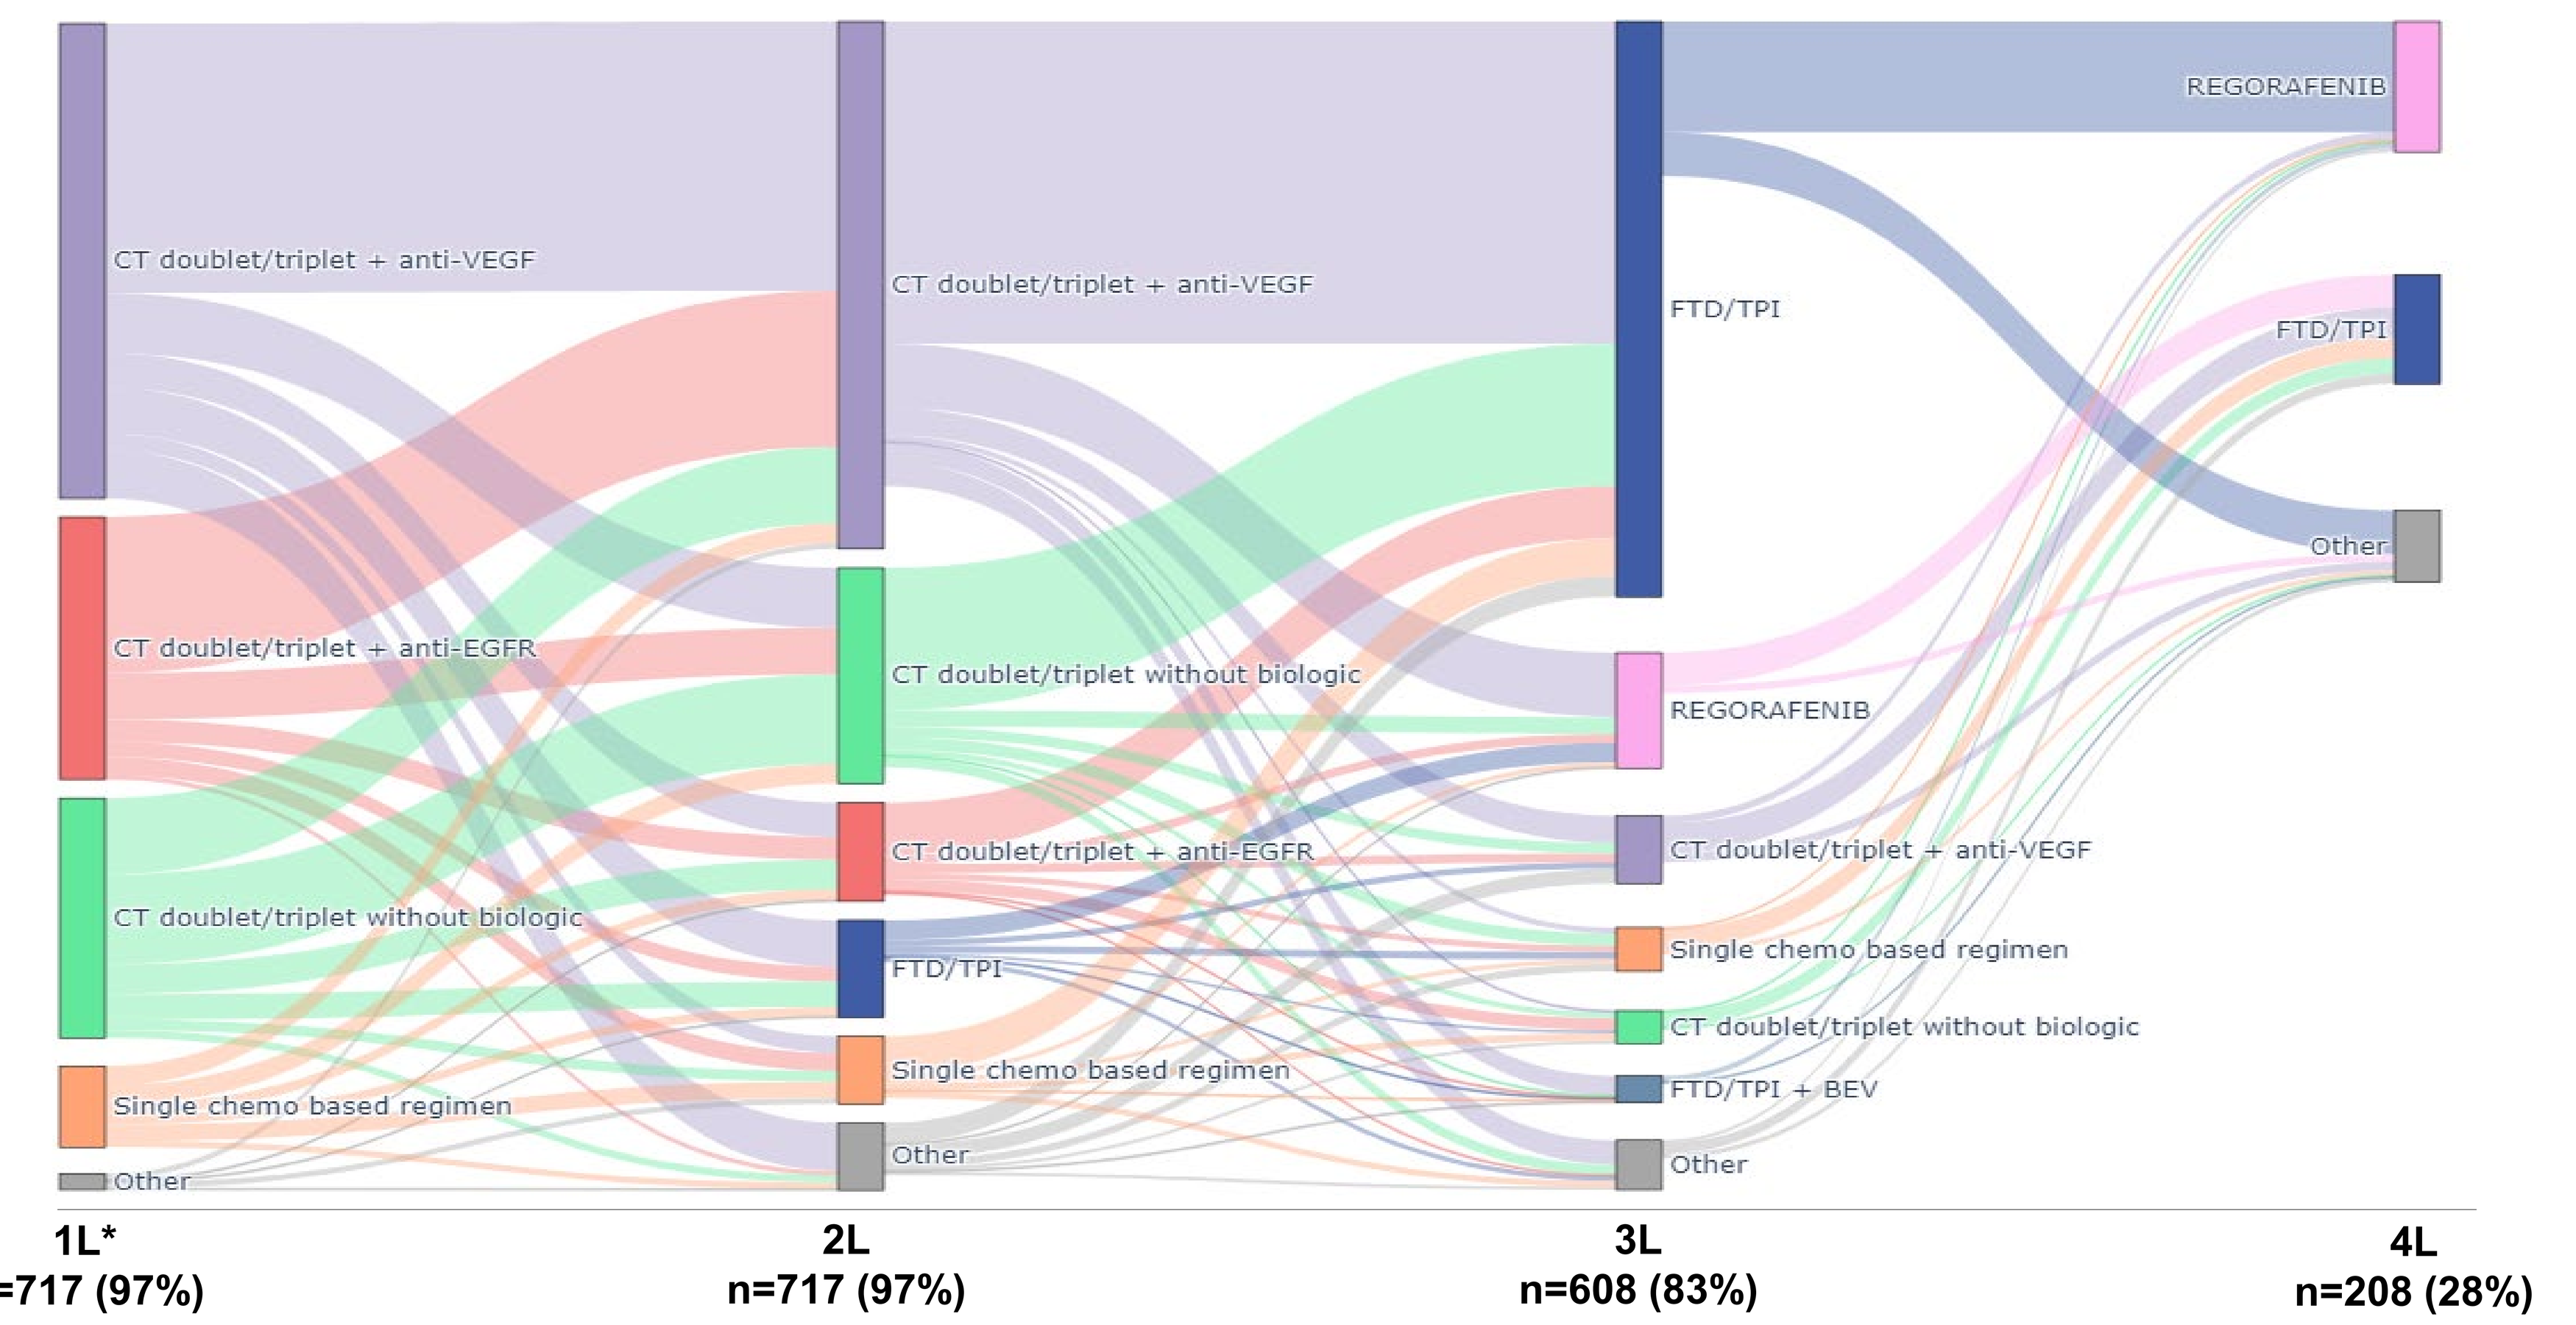
CT doublet/triplet + anti-VEGF
CT doublet/triplet + anti-EGFR
CT doublet/triplet without biologic
Single chemo based regimen
Other
CT doublet/triplet + anti-VEGF
CT doublet/triplet without biologic
CT doublet/triplet + anti-EGFR
FTD/TPI
Single chemo based regimen
Other
FTD/TPI
REGORAFENIB
CT doublet/triplet + anti-VEGF
Single chemo based regimen
CT doublet/triplet without biologic
FTD/TPI + BEV
Other
REGORAFENIB
FTD/TPI
Other
1L*
n=717 (97%)
2L
n=717 (97%)
3L
n=608 (83%)
4L
n=208 (28%)

*Analysis of 717/736 patients with ≥2 LoTs. Other: grouping treatment categories with fewer than 10 patients. Abbreviations: FTD/TPI + BEV, trifluridine/tipiracil + bevacizumab; n, number; CT, chemotherapy; VEGF, vascular endothelial growth factor; EGFR, epidermal growth factor receptor; 1L, first-line; 2L, second-line; 3L, third-line; 4L, fourth-line.

Overall survival

- Use of CT and biologic agents in 1L and 2L, followed by FTD/TPI ± bevacizumab in 3L, led to a mOS from 1L of >23 months, and mOS was lower when a biologic was not used in 1L/2L (**Table 1**).
- In patients who received 4L treatment, most patients received CT and biologics in 1L/2L, 3L FTD/TPI ± bevacizumab, and 4L regorafenib, and these patients had a mOS of 29.9 months (anti-vascular endothelial growth factor (VEGF) twice in 1L/2L) and 34.7 months (anti-VEGF + anti-epidermal growth factor receptor (EGFR) in 1L/2L) (**Table 1**).

Table 1: Overall survival by treatment sequence from first-line in patients who received 3 or 4 lines of treatment

Treatment pathway	Patients (n)	mOS (95% CI)
Treatment pathway in patients with 3 lines of therapy		
1L/2L doublet/triplet CT + **anti-VEGF** → 3L **FTD/TPI ± BEV**	90	23.8 (21.6–28.6)
1L/2L CT + **anti-EGFR**/CT + **anti-VEGF** (any order) → 3L **FTD/TPI ± BEV**	60	28.6 (26.4–37.9)
1L/2L doublet/triplet CT without biologic → 3L **FTD/TPI ± BEV**	40	20.6 (15.7–24.7)
1L/2L/3L at least one immunotherapy / targeted therapy	13	23.8 (20.4–NR)
Doublet/triplet CT + **anti-VEGF** only once → **FTD/TPI ± BEV** (2L or 3L) after 'doublet/triplet CT + anti-VEGF' And not in groups 1-4	62	34.8 (27.2-43.9)
All other treatment pathways with at least three LoTs	135	30.7 (26.6–35.9)
Treatment pathway in patients with 4 lines of therapy		
1L/2L doublet/triplet CT + **anti-VEGF** → 3L **FTD/TPI ± BEV** → 4L **regorafenib**	35	29.9 (24.0–43.1)
1L/2L CT + **anti-EGFR**/CT + **anti-VEGF** (any order) → 3L **FTD/TPI ± BEV** → 4L **regorafenib**	16	34.7 (31.0–40.9)
1L/2L/3L/4L at least one **immunotherapy / targeted therapy**	9	26.8 (16.5–NR)
Doublet/triplet CT + **anti-VEGF** prior to '**FTD/TPI** (at 2L, 3L, or 4L)' Not included in groups 6–8	48	39.7 (31.9–46.9)
Any patient who received four LoTs and is not included in groups 6–9	58	33.9 (31.6–38.4)

Abbreviations: mOS, median overall survival; CI, confidence interval; FTD/TPI ± BEV, trifluridine/tipiracil + bevacizumab; n, number; CT chemotherapy; VEGF vascular endothelial growth factor; EGFR, epidermal growth factor receptor; NR, not reached; 1L, first-line; 2L, second-line; 3L, third-line; 4L, fourth-line.

Focus on patients treated with FTD/TPI + bevacizumab

- 23 patients were treated with FTD/TPI + bevacizumab in the PROMETCO study, and of these, 1 patient received FTD/TPI + bevacizumab as 2L therapy, 18 patients received it as 3L therapy, and 5 patients received it as 4L therapy.
- Due to the timing of the study only 4.1% of patients received FTD/TPI + bevacizumab, however efficacy outcomes in these patients were consistent with the SUNLIGHT trial.[3]
- Median (95% confidence interval [CI]) progression-free survival and OS from diagnosis was 2.89 (1.64–3.78) months and 39.1 (27.2–75.1) months, respectively. Median OS (95% CI) from PROMETCO inclusion was 13.6 (7.43-21.2) months.

Multivariable analysis

- Being fitter and having a lower disease burden (Eastern Cooperative Oncology Group Performance Status (ECOG PS) 0-1, good prognosis characteristics (GPCs), and Microsatellite Stability (MSS)) led to a significantly increased OS after two progressions of disease (**Table 2**).
- Other factors that led to significantly improved OS from PROMETCO inclusion were having surgery and being more than 65 years old (**Table 2**). Time since diagnosis, and tumor sidedness (left or right) did not influence OS from PROMETCO inclusion.

Table 2: Multivariable analysis of factors influencing overall survival from PROMETCO inclusion

Characteristic	n (%)	HR (multivariable) (95% CI, p)
Age		
<65 years	223 (40.0)	-
≥65 years	335 (60.0)	0.71 (0.56-0.90, p=0.005)
ECOG PS		
0/1	504 (92.6)	-
≥2	40 (7.4)	2.31 (1.53-3.47, p<0.001)
Prognostic sub-group*		
GPC	330 (59.1)	-
PPC	228 (40.9)	1.68 (1.30-2.16, p<0.001)
MSI status		
MSI-high	9 (2.7)	-
MSS	326 (97.3)	0.39 (0.19-0.79, p=0.009)
Surgery		
No	185 (33.2)	-
Yes**	373 (66.8)	0.64 (0.50-0.83, p=0.001)

Abbreviations: HR, hazard ratio; MSI, microsatellite instability; MSS, microsatellite stable; ECOG PS, eastern cooperative oncology group performance status; GPC, good prognosis characteristics; PPC, poor prognosis characteristics; CI, confidence interval.
*Good prognosis characteristics [GPC], defined as having <3 metastatic sites at study entry and ≥18 months from diagnosis of metastatic disease to study entry, best prognosis characteristics and the remaining patients had poor prognosis characteristics [PPC].[6] **At least one colorectal, liver or lung surgery

- For OS from 1L, as expected, having surgery, being ECOG PS 0-1, and having GPCs were positive prognostic factors and significantly increased OS (**Table 3**).
- Having a longer time since diagnosis positively and significantly impacted OS from 1L but not after two progressions of disease.

Table 3: Multivariable analysis of factors influencing overall survival from first-line

Characteristic	n (%)	HR (multivariable) (95% CI, p)
Time since diagnosis at baseline (5 months change), months	6.5 (5.0)+	0.85 (0.82-0.87, p<0.001)
ECOG PS		
0/1	504 (92.6)	-
≥2	40 (7.4)	2.22 (1.58-3.12, p<0.001)
Prognostic sub-group*		
GPC	330 (59.1)	-
PPC	228 (40.9)	4.73 (3.62-6.18, p<0.001)
Surgery		
No	185 (33.2)	-
Yes**	373 (66.8)	0.74 (0.60-0.91, p=0.005)

Abbreviations: HR, hazard ratio; MSI, microsatellite instability; MSS, microsatellite stable; ECOG PS, eastern cooperative oncology group performance status; GPC, good prognosis characteristics; PPC, poor prognosis characteristics; CI, confidence interval.
+Reported as mean (SD), not n (%); *Good prognosis characteristics [GPC], defined as having <3 metastatic sites at study entry and ≥18 months from diagnosis of metastatic disease to study entry, best prognosis characteristics and the remaining patients had poor prognosis characteristics [PPC].[6]; **At least one colorectal, liver or lung surgery

References

1. Taieb J, Price T, Vidot L, et al. BMC Cancer. 2023;23(1): 94.
2. Weiss L, Karthaus M, Riera-Knorrenschild J, et al. ESMO Open. 2022;7(1): 100391.
3. Prager GW, Taieb J, Fakih M, et al. N Engl J Med. 2023;388(18):1657-67.
4. Dasari A, Lonardi S, Garcia-Carbonero R, et al. The Lancet. 2023;402(10395): 41-53.
5. Koopman M, Pinto C, Bodoky G, et al. Future Oncol. 2022;18(11):1313-1320.
6. Tabernero J, Argiles G, Sobrero AF, et al. ESMO Open. 2020;5(4).

Acknowledgements

This study is sponsored by Servier Affaires Médicales, France.
Editorial assistance was provided by Emily Eagles of ESP Medical Communications Ltd, and supported by Servier.

Corresponding Author & Disclosures

Corresponding email: jean-baptiste.bachet@aphp.fr

Disclosures: **JBB**: Received personal fees from Abbvie, Amgen, Bayer, Bristol Myers Squibb, GlaxoSmithKline, Merck Serono, Merck Sharp & Dohme, Pierre Fabre, Sanofi, Servier, and Takeda; **KM** : Honoraria from Servier; **CP**: Received honoraria for acting as an advisory board member or speaker for Amgen, Astellas, AstraZeneca, Bayer, BMS, Daiichi Sankyo, Janssen, Lilly, Merck Serono, MSD, Novartis, Pfizer, Roche, Sandoz, Sanofi and Servier; **RGC**: has provided scientific advice and/or received honoraria or funding for continuous medical education from AAA-Novartis, Advanz Pharma, Amgen, Astellas, Bayer, BMS, Boehringer, Crinetics, Esteve, GSK, Hutchmed, Ipsen, ITM, MSD, PharmaMar, Pierre Fabre, Sanofi, Servier, Takeda; has received research grants from BMS, MSD, and Pfizer, has Leadership Role, Global PI of investigator-initiated clinical trials (AXINET, NICENEC, PEMBROLA): BMS, MSD, Pfizer, other - Honoraria received by spouse for advisory board or invited speaker roles: AbbVie, AstraZeneca, Genomica, Lilly, Merck, Pfizer, Roche, Sanofi; **GB**: Received honoraria as an advisory board member or speaker from Amgen, Astra Zeneca, Roche, Bayer, Pfizer, Janssen, Novartis, Lilly, Servier, Nordic Pharma, Taiho; **AM:** Received honoraria or grants from AstraZeneca, Bayer, Lilly, Servier, Pierre Fabre and Merck; **AS, EC, BC**: Are employees of Servier; **MK**: Reports having an advisory role for Eisai, Nordic Farma, Merck-Serono, Pierre Fabre, Servier (paid to institution), Institutional scientific grants from Bayer, Bristol Myers Squibb, Merck, Personal Genome Diagnostics (PGDx), Pierre Fabre, Roche, Sirtex, Servier.